Daptomycin Susceptibility of *Staphylococcus aureus* Isolates Causing Infections in Australia.

J Pearson[1], J Bell[2], G Coombs[1], S Cramer[1], J Fox[3].

[1]PathWest Laboratory Medicine – WA Royal Perth Hospital, Western Australia.
[2]SA Pathology, Women's and Children's Hospital, South Australia.
[3]Novartis Pharmaceuticals Australia Pty Ltd, New South Wales, Australia.

Background

Daptomycin is a cyclic lipopeptide antimicrobial with activity against Gram-positive bacteria. It has a novel mechanism of action; utilizing calcium ions to bind irreversibly to the cell membranes causing rapid depolarization of membrane potential resulting in efflux of potassium and destruction of the ion-concentration gradient. Loss of membrane potential leads to inhibition of protein, DNA, and RNA synthesis, resulting in bacterial cell death. It is approved for use in treatment of patients with complicated skin and soft tissue infection and *Staphylococcus aureus* bacteraemia including right-side native value endocarditis.

Objectives

To determine minimum inhibitory concentration (MIC) of *S. aureus* to daptomycin in an Australian outpatient population.

Methods

- The Australian Group for Antimicrobial Resistance (AGAR) collected 3,075 clinically significant *S. aureus* isolates from outpatients in 2008.
- Isolates were from skin and soft tissue infections (88%), respiratory (5%), urine (2%), blood (2%), eye (2%) sterile body cavity (<1%), ear (<1%) and not specified (<1%).
- Daptomycin MIC was determined by Etest (AB Biodisk, Sweden) using BBL™ II Mueller Hinton agar (batch 811856). MIC was recorded where the inhibition ellipse intersected the MIC scale.
- Twenty seven isolates with an MIC >1 mg/L (non-susceptible) were re-tested by Etest at a central laboratory.
- Six of these isolates were tested by broth microdilution using CLSI guidelines.

Results

- After initial testing at the referring laboratories, 28 (0.9%) isolates had an MIC in the non-susceptible range (MIC >1mg/L) by CLSI and EUCAST breakpoints.
- Re-testing of isolates at a central laboratory resulted in lower MICs, often 2 to 3 doubling dilutions lower than that recorded at the referring laboratory (Table 1).
- One isolate had an MIC of 1.5mg/L both at the referring and central laboratory. The MIC of this isolate was 1mg/L by broth microdilution.
- A further 5 isolates of varying MICs were confirmed as susceptible by broth microdilution.
- After repeat testing all 3,075 *S. aureus* isolates tested as susceptible to daptomycin.
- The MIC_{50} was 0.25mg/L and the MIC_{90} 0.38mg/L.
- A slight movement when placing the Etest strip on the media resulted in discrepant MICs on either side of the strip (Figure 1).

Table 1: Etest MIC (mg/L) comparison between referring lab and central laboratory (n)

Referring Lab MIC / Central Lab Repeat MIC	1.5	2	3
0.094		1	
0.125	6	4	1
0.19	5	5	
0.25	1	1	1
0.38			
0.5			
0.75	2		
1			
1.5	1*		

* MIC 1mg/L by broth microdilution

Figure 1: Discrepant MIC on left and right sides of Etest strip following slight movement of strip when placing on the media

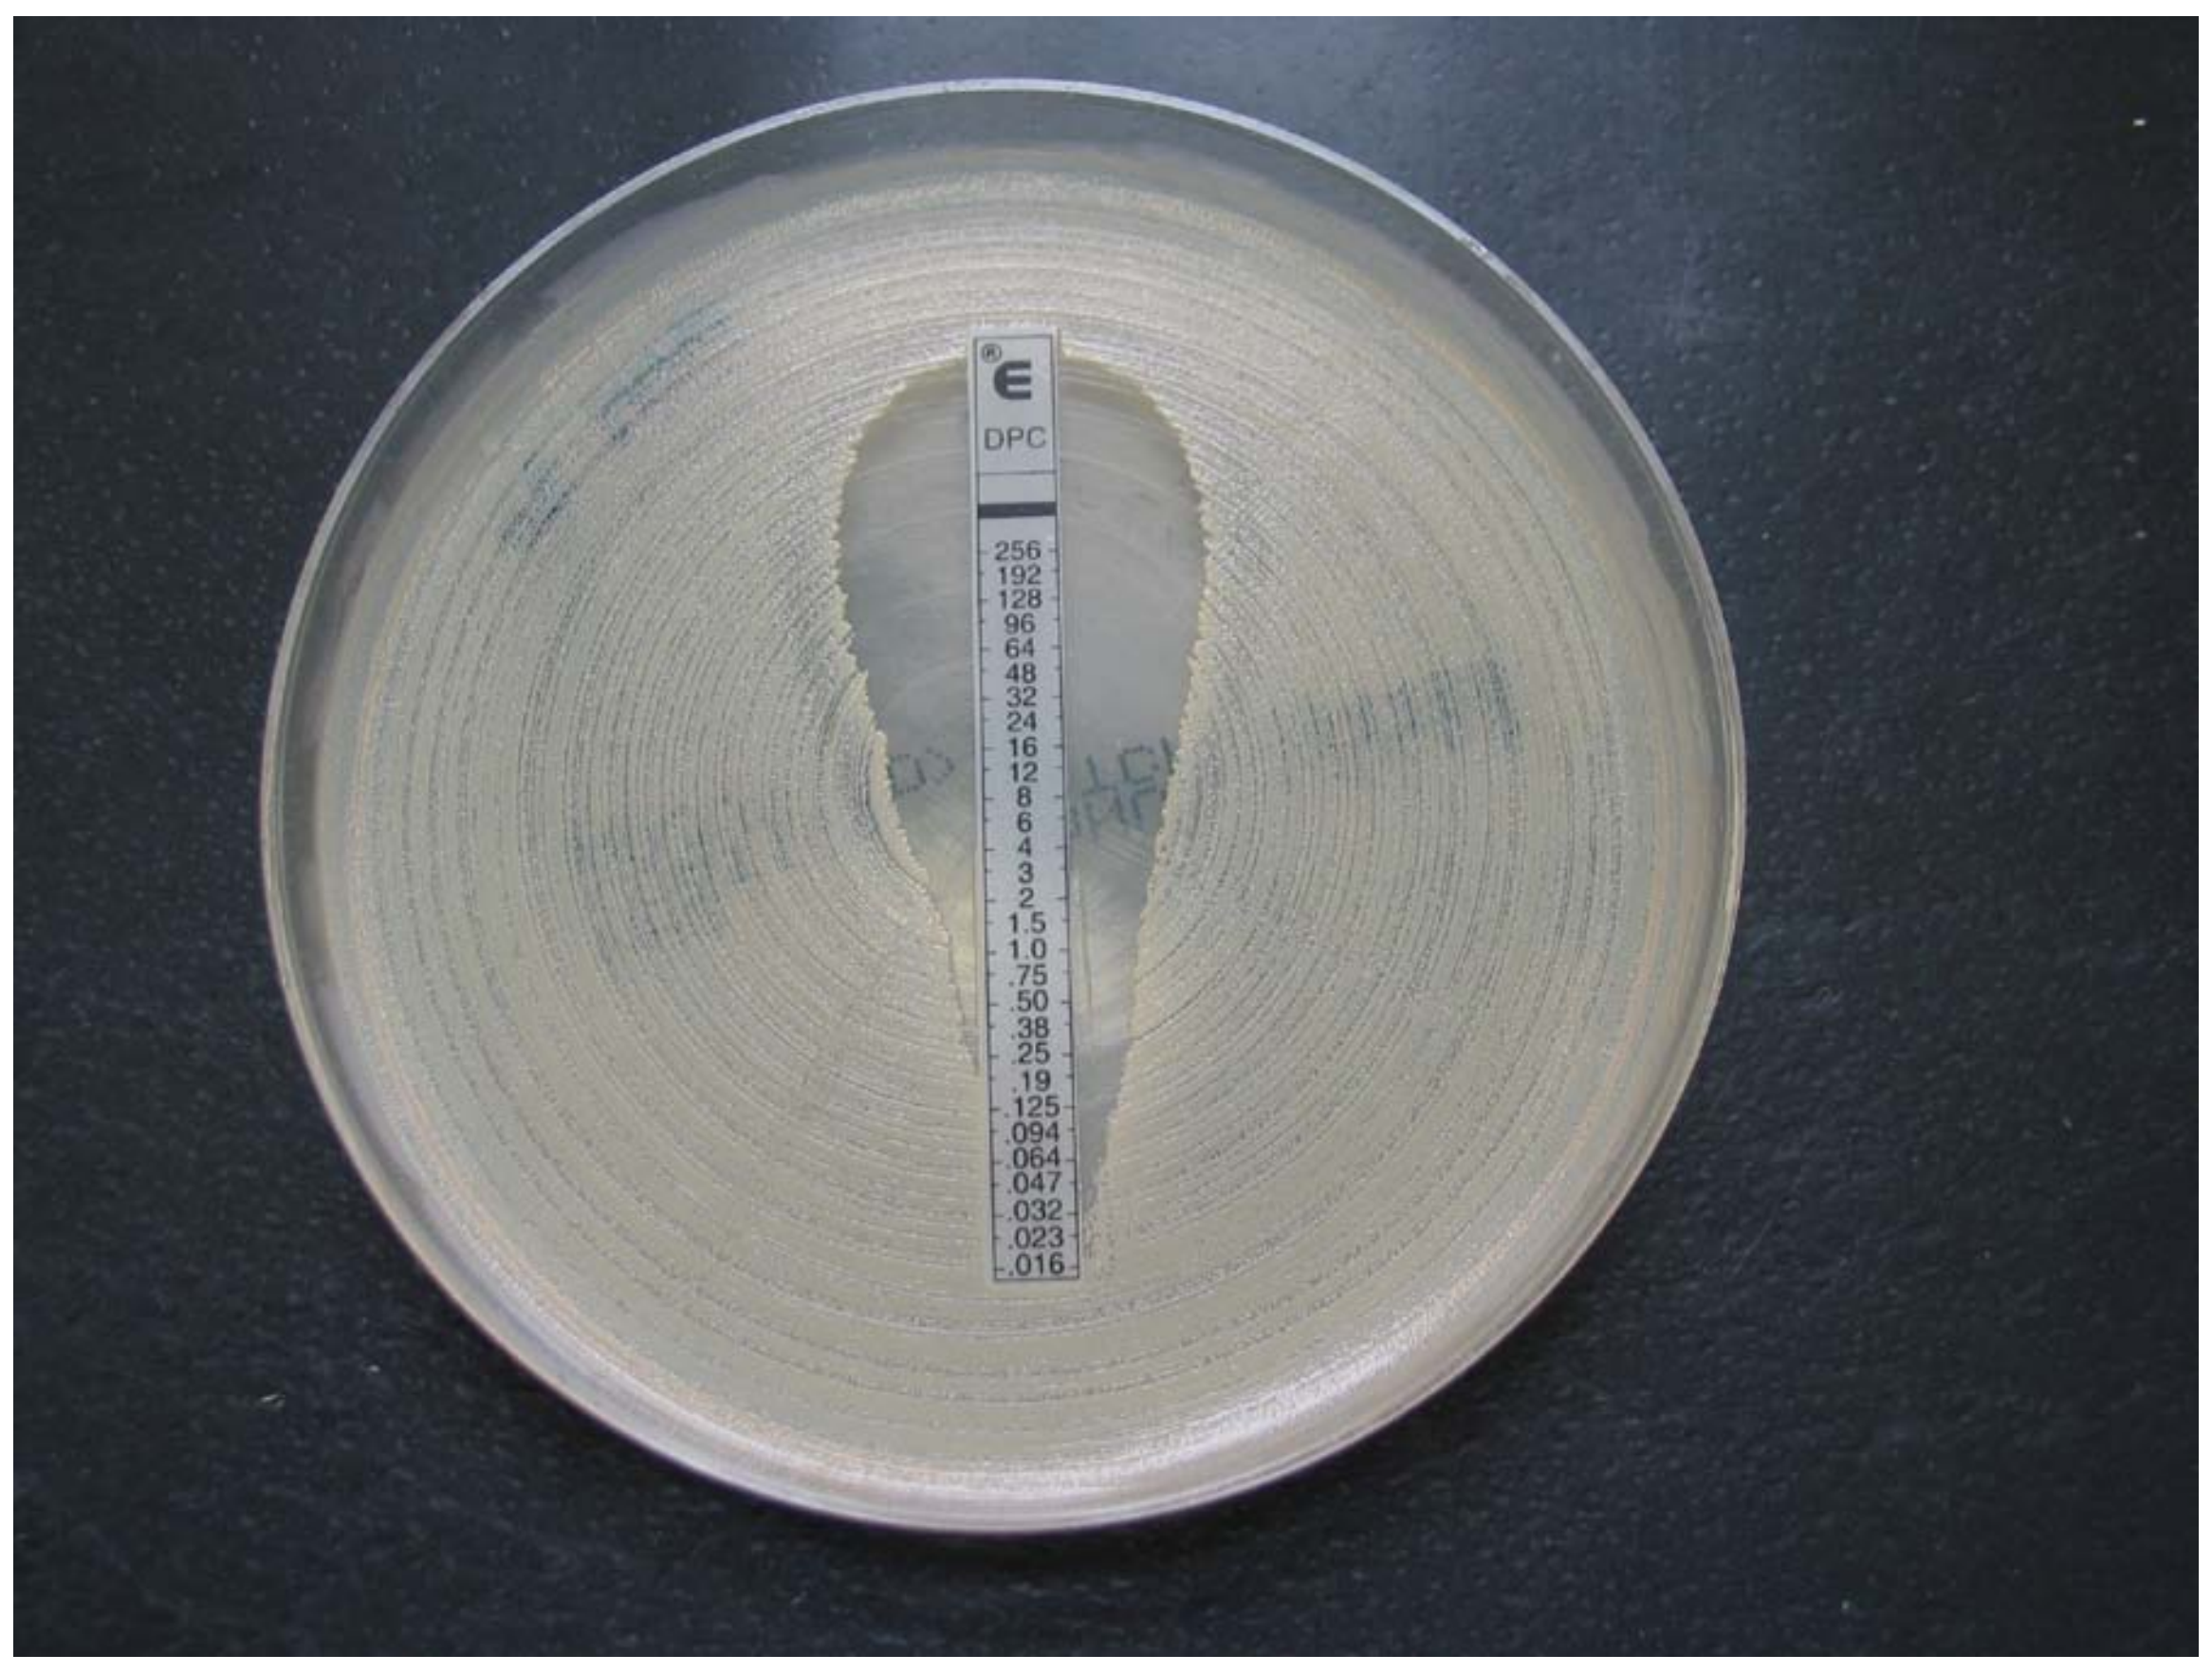
DPC
256
192
128
96
64
48
32
24
16
12
8
6
4
3
2
1.5
1.0
.75
.50
.38
.25
.19
.125
.094
.064
.047
.032
.023
.016

Conclusions

- The high MICs (>1mg/L) recorded at initial testing may have been due to the recording of the higher MIC where a discrepancy was evident between both sides of the strip.
- An MIC result of >1mg/L should be treated with caution as daptomycin non-susceptibility was not detected in this survey and has been reported infrequently by other surveillance programmes (SENTRY, BSAC, HPA UK).
- Testing should be repeated on isolates with a MIC >1mg/L taking care when placing the Etest strip on to the media. If result is duplicated, the isolate should be referred for MIC determination using broth microdilution.

Acknowledgements

The following institutions participated in this survey: Alfred Hospital VIC, Austin Health VIC, Concord Hospital NSW, Douglass Hanly Moir Pathology NSW, Gribbles Pathology VIC, John Hunter Hospital NSW, Launceston General Hospital TAS, Monash Medical Centre VIC, Nepean Hospital NSW, Pathology Queensland Cairns Base Hospital QLD, Pathology Queensland Central Laboratory QLD, Pathology Queensland Gold Coast Hospital QLD, Pathology Queensland Princess Alexandra Hospital QLD, Pathology Queensland Prince Charles Hospital QLD, PathWest Laboratory Medicine Fremantle Hospital WA, PathWest Laboratory Medicine Queen Elizabeth II Medical Centre WA, PathWest Laboratory Medicine Royal Perth Hospital WA, Royal Women's' and Children's Hospitals VIC, Royal Darwin Hospital NT, Royal Hobart Hospital TAS, Royal North Shore Hospital NSW, Royal Prince Alfred Hospital NSW, SA Pathology Flinders Medical Centre SA, SA Pathology IMVS SA, SA Pathology Women's and Children's Hospital SA, St John of God Pathology WA, Sullivan and Nicolaides Pathology QLD, St Vincent's Hospital VIC, Sydney South West Area Pathology NSW, The Canberra Hospital ACT, Westmead Hospital NSW

AGAR is partially funded by the Department of Health and Ageing. Novartis provided partial funding and the consumables for this study.

julie.pearson@health.wa.gov.au